The Use of Remote Centralized Raters via Live Two-Way Video in a Multicenter Clinical Trial for Schizophrenia

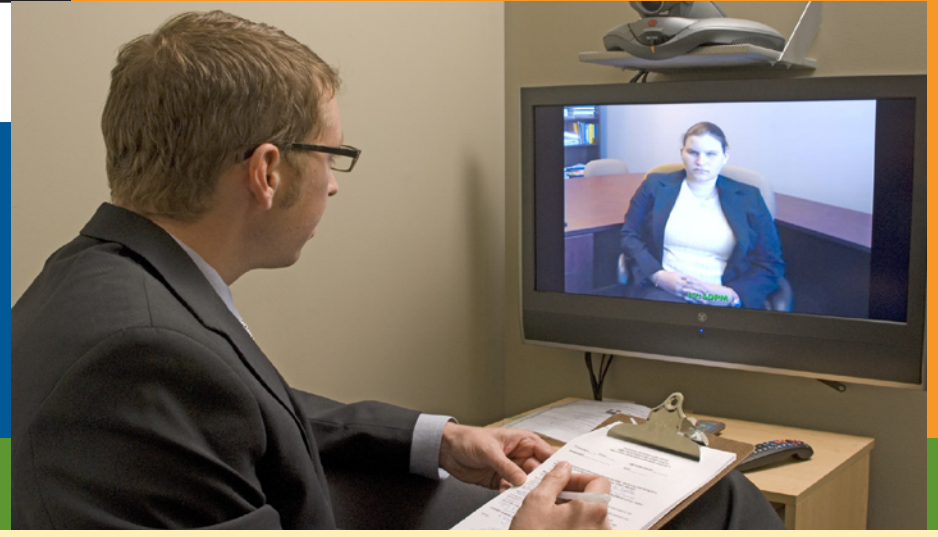

Joan Shen, MD, PhD[1] Kenneth A. Kobak, PhD[2] Yonggang Zhao, PhD[1] Madeline G. Alexander PhD[2] John M.Kane, MD[3]

[1] Wyeth Research [2] MedAvante, Inc. [3] The Zucker Hillside Hospital and The Albert Einstein College of Medicine

Background: The growing rate of failed and negative CNS trials has led to increased examination of clinical trial methodology. Factors associated with clinician assessment, such as expectancy bias, rating inflation due to enrollment pressure, poor inter-rater reliability, and poor interview quality, may play a role in the failure of marketed and truly effective novel compounds to separate from placebo. One proposed solution has been the use of centralized raters, i.e., a small group of highly skilled, tightly calibrated, and continuously monitored raters who are linked to the various study sites through videoconferencing. The use of centralized raters can improve reliability by reducing the sheer number of raters involved (e.g., 8-10 raters vs. 60-75). They can be rigorously calibrated using procedures not logistically feasible with a larger group of raters at diffuse study sites. Being divorced from the study site and blind to inclusion criteria eliminates the pressure to enroll patients or inflate baseline scores. Blinding to study visit reduces expectancy bias. The use of centralized raters in schizophrenia trials raises several issues, such as whether acutely psychotic patients would be willing or able to participate, and whether raters using videoconferencing would be able to find a significant drug effect. The current phase II study is the first of several RCTs to use centralized raters in a study of treatments for schizophrenia.

Methods: Subjects (N=313) from 32 sites, aged 18-65, with an acute exacerbation of schizophrenia were randomly assigned to 6 weeks of treatment with one of two doses of an experimental compound, active comparator (olanzapine), or placebo. Subjects were evaluated weekly using the Positive and Negative Syndrome Scale (PANSS) by a centralized rater who was blinded to the study visit and protocol, and provided with data on the patient's behavior from an informant who observed the patient during the previous week.

A different rater typically saw the patient each visit. Data from the olanzapine (N=68) and placebo (N=68) arms were provided by the sponsor.

Results from the central raters' PANSS interviews were faxed back to the site within 30 minutes. The site principal investigator managed the patient clinically and was responsible for all the study related clinical and protocol decisions. While the site (not the central) rater was responsible for monitoring adverse events, if any were disclosed during the central raters' PANSS interview, the central rater forwarded this information to the sites. The sites then followed-up with the patient if necessary for clinical management and to make a determination regarding classifying this information.

Subjects were scheduled as usual by the sites, who informed the central raters of the date and time of the next visit. Eight percent of assessments were conducted on the same day they were scheduled, 34% within 2 days of scheduling, and 58% with 3 or more days' advance notice. The site principal investigator managed the patient clinically and was responsible for all study-related clinical and protocol decisions.

Videoconference interviews were conducted via T1 line over a virtual private network (VPN), run at a minimum of an industry standard bit rate of 384kps guaranteeing both availability and high bandwidth, insuring speed and picture quality.

The central raters (N=18) all had experience working with patients with schizophrenia (mean=8.4 years), and administering the PANSS (mean=4.3 years). Twelve raters (66%) had doctoral degrees and 6 (33%) had masters degrees. All raters went through a rigorous training and calibration procedure (ICC=.91). ICC was monitored at regular intervals throughout the study.

Results: A significant difference in change from baseline in PANSS total score was found between olanzapine and placebo starting at week 1, which continued throughout the study. At endpoint, the adjusted mean change seen with olanzapine (15.2 points, SE=2.52) was significantly greater than the mean change for placebo (4.43 points, SE=2.51), p=.002 from an analysis of covariance (ANCOVA) model with last observation carried forward (LOCF). A significant difference (p=.021) was also observed from the mixed model with observed cases. The effect size at endpoint was .48. Internal consistency was high throughout the study (.92 and .89 at endpoint for olanzapine and placebo, respectively). Scores at screening were normally distributed and not skewed toward the cutoff score. Overall, 1993 remote assessments were completed during the 13-month study. Of the 1993 assessments, 39 (2%) experienced temporary interruptions because of technical issues, which were immediately resolved, and the interviews were completed. In 10 cases (0.5%) the interview could not be completed and had to be rescheduled.

Discussion: Results indicate that acutely psychotic patients with schizophrenia are willing and able to participate in clinical trials using remote interviews conducted via videoconference. Internal consistency and reliability were high, and scores did not appear to be inflated at baseline by blinded raters. This in itself could reduce the placebo response that has been seen in recent trials. Centralized raters detected significant differences between active drug and placebo, with an effect size similar to or better than those reported in the literature for similar doses with the active comparator. This methodology shows enormous promise for use in clinical trials.

Figure 1.
Adjusted Mean Change in PANSS Total Score by Study Week (ANCOVA)

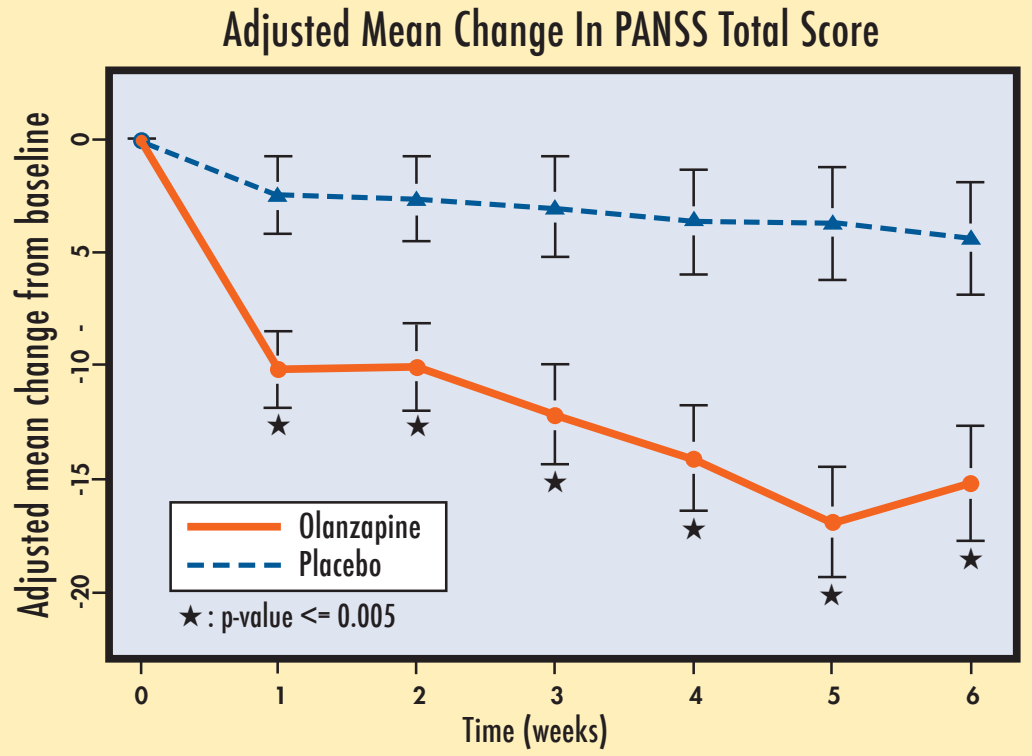
Adjusted Mean Change In PANSS Total Score
Adjusted mean change from baseline
0
5
10
15
20
Olanzapine
Placebo
★ : p-value <= 0.005
0
1
2
3
4
5
6
Time (weeks)

Frequency Distribution: Screening PANSS Scores
VISIT: 1

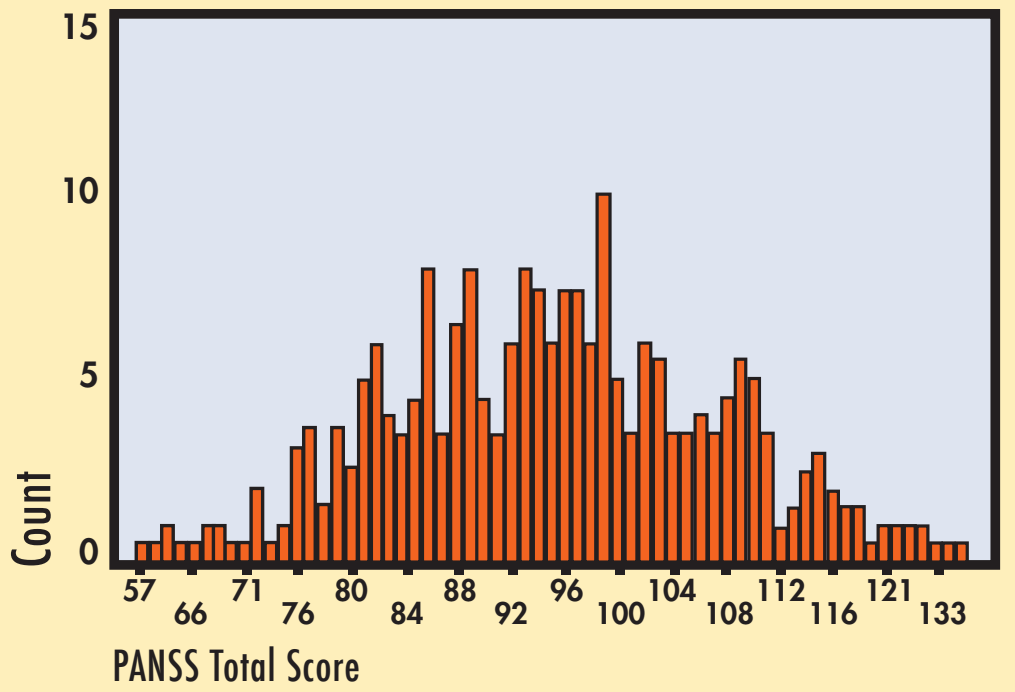
15
10
5
0
Count
57
66
71
76
80
84
88
92
96
100
104
108
112
116
121
133
PANSS Total Score

Mean Change, PANSS

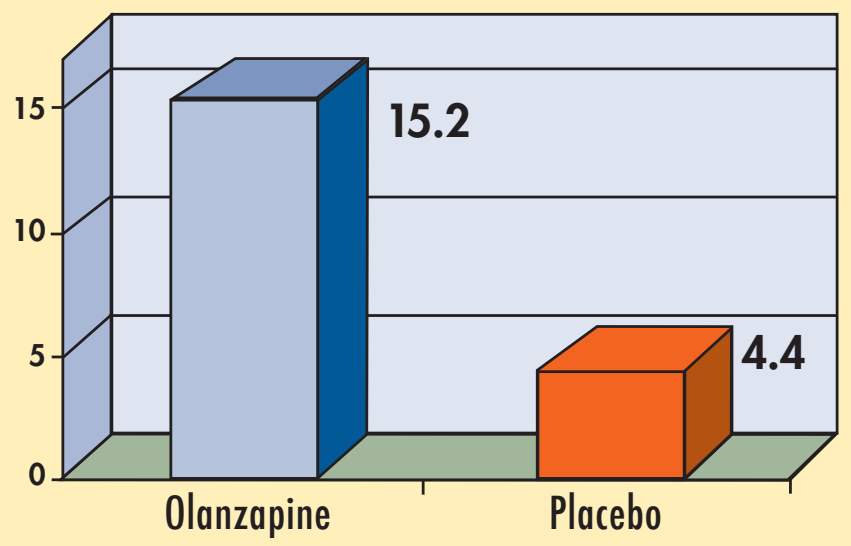
15
10
5
0
15.2
4.4
Olanzapine
Placebo

MEDAVANTE® RESEARCH INSTITUTE
Subjective -> Objective

MedAvante Inc., 100 American Metro Boulevard, Suite 106, Hamilton, NJ 08619
© MedAvante, Inc. 2008